P2048

BEST RESPONDERS TO CALCIPOTRIOL/BETAMETHASONE DIPROPIONATE (CAL/BDP) PAD-CREAM: FIVE CASE REPORTS OF PATIENTS WITH MILD-TO-MODERATE PLAQUE PSORIASIS

Andreas Pinter[1], Jordi Galván[2], Frank Freischläger[3]

[1]Department of Dermatology, University Hospital Frankfurt am Main, Frankfurt, Germany
[2]Global Medical Affairs Department, Almirall S.A., Barcelona, Spain
[3]Estimondo GmbH, Nortmoor, Germany

INTRODUCTION

- A calcipotriol (CAL) and betamethasone dipropionate (BDP) cream based on polyaphron dispersion (PAD) technology emerged as a novel formulation for a more convenient topical treatment of psoriasis.[1]
- Individual endpoints from phase 3 trials have demonstrated high efficacy, a favourable safety and tolerability profile, and convenience for CAL/BDP PAD-cream for the treatment of adults with mild-to-moderate psoriasis.[2]
- Recently, a *post-hoc* pooled analysis of the two phase 3 trials showed that, after the recommended 8-week treatment period, more than one in four patients treated with CAL/BDP PAD-cream achieved the best response composite endpoint (defined in Materials & Methods). When compared to CAL/BDP gel, the active comparator in the phase 3 trials, the result was statistically significant, with a higher best response rate observed for CAL/BDP PAD-cream than CAL/BDP gel (27.4% vs. 16.1%; p<0.0001) at Week 8.[3]

OBJECTIVES

- The aim of this analysis is to provide further insights into the demographic characteristics and outcomes of some best responder patients to CAL/BDP PAD-cream.

MATERIALS AND METHODS

Study design and patients

- We report five cases of adult patients with mild-to-moderate plaque psoriasis from the previously published multicenter, randomized, investigator-blinded, active and vehicle-controlled, phase 3 trial MC2-01-C7 (NCT03802344).[4]
- All patients applied CAL/BDP PAD-cream to the affected areas once daily for 8 weeks.
- Patients were included if they had photographs taken at baseline and at each study visit, and if they achieved the composite endpoint of best response at Week 4 and/or Week 8.

Best response definition[3]

- Best responders were defined as patients achieving at Week 4 and/or Week 8 a restrictive composite endpoint made by the combination of:
 - ✓ Physician's Global Assessment (PGA) controlled disease: any improvement in PGA to score of 0-1 from baseline

 +
 - ✓ Modified Psoriasis Area and Severity Index (mPASI) success: mPASI <2

 +
 - ✓ Dermatology Life Quality Index (DLQI) satisfaction: DLQI 0-1.

Data collection and outcomes

- For each patient, baseline demographics and disease characteristics were recorded.
- Additionally, PGA, mPASI and DLQI scores were collected at baseline, Week 4 and Week 8. Only observed values were included in the analyses. No imputation of missing data was performed.
- Photographic documentation of plaque psoriasis progression at different locations was also obtained from baseline through Week 8.

RESULTS

Patients

- Five male patients with mild-to-moderate psoriasis were analysed, with a mean age of 56.2 years, a mean body mass index (BMI) of 27.3 kg/m^2 and a mean psoriasis duration of 18.0 years.
- Individual baseline demographic and disease characteristics are shown in **Table 1**.

Table 1. Baseline demographic and disease characteristics

Case Number	Age (years)	Sex	BMI (kg/m^2)	Race	Ethnic Origin	Psoriasis duration (years)	Fitzpatrick skin type
Case 1	68	Male	33.4	White	Not Hispanic or Latino	20.7	Type II
Case 2	65	Male	31.2	White	Not Hispanic or Latino	25.6	Type II
Case 3	55	Male	27.2	White	Not Hispanic or Latino	16.6	Type II
Case 4	59	Male	22.5	White	Not Hispanic or Latino	20.0	Type II
Case 5	34	Male	22.3	White	Not Hispanic or Latino	6.9	Type II

BMI, body mass index.

Best response to treatment

- PGA, mPASI, and DLQI results per patient at baseline, Week 4 and Week 8 are shown in **Table 2**.
- Although all patients were best responders at Week 8, one of them had already reached best response at Week 4. Case 4 demonstrated an early response to the composite endpoint, as patient achieved a PGA score of 0-1, an mPASI <2 and a DLQI score of 0-1 by Week 4.

Table 2. PGA, mPASI and DLQI at baseline, Week 4 and Week 8

Case Number	PGA			mPASI			DLQI		
	Baseline	Week 4	Week 8	Baseline	Week 4	Week 8	Baseline	Week 4	Week 8
Case 1	3	1	1	5.4	2.2	1	11	0	0
Case 2	3	2	1	3.6	1.8	0.4	1	0	1
Case 3	3	2	0	7.2	4.8	0	11	4	0
Case 4	3	1	0	4.8	1.8	0	10	0	0
Case 5	3	2	1	7.8	1.4	1.5	5	2	1

PGA, Physician's Global Assessment; mPASI, modified Psoriasis Area and Severity Index; DLQI, Dermatology Life Quality Index.

Visual evolution of psoriasis

- Images illustrating the progression of clinical outcomes after 4 and 8 weeks of treatment with CAL/BDP PAD-cream are presented in **Figure 1**.

Figure 1. Evolution of psoriasis during 8 weeks of CAL/BDP PAD-cream treatment

CASE 1

Knees: Near-complete clearance of erythemato-squamous plaques already observed after 4 weeks of treatment

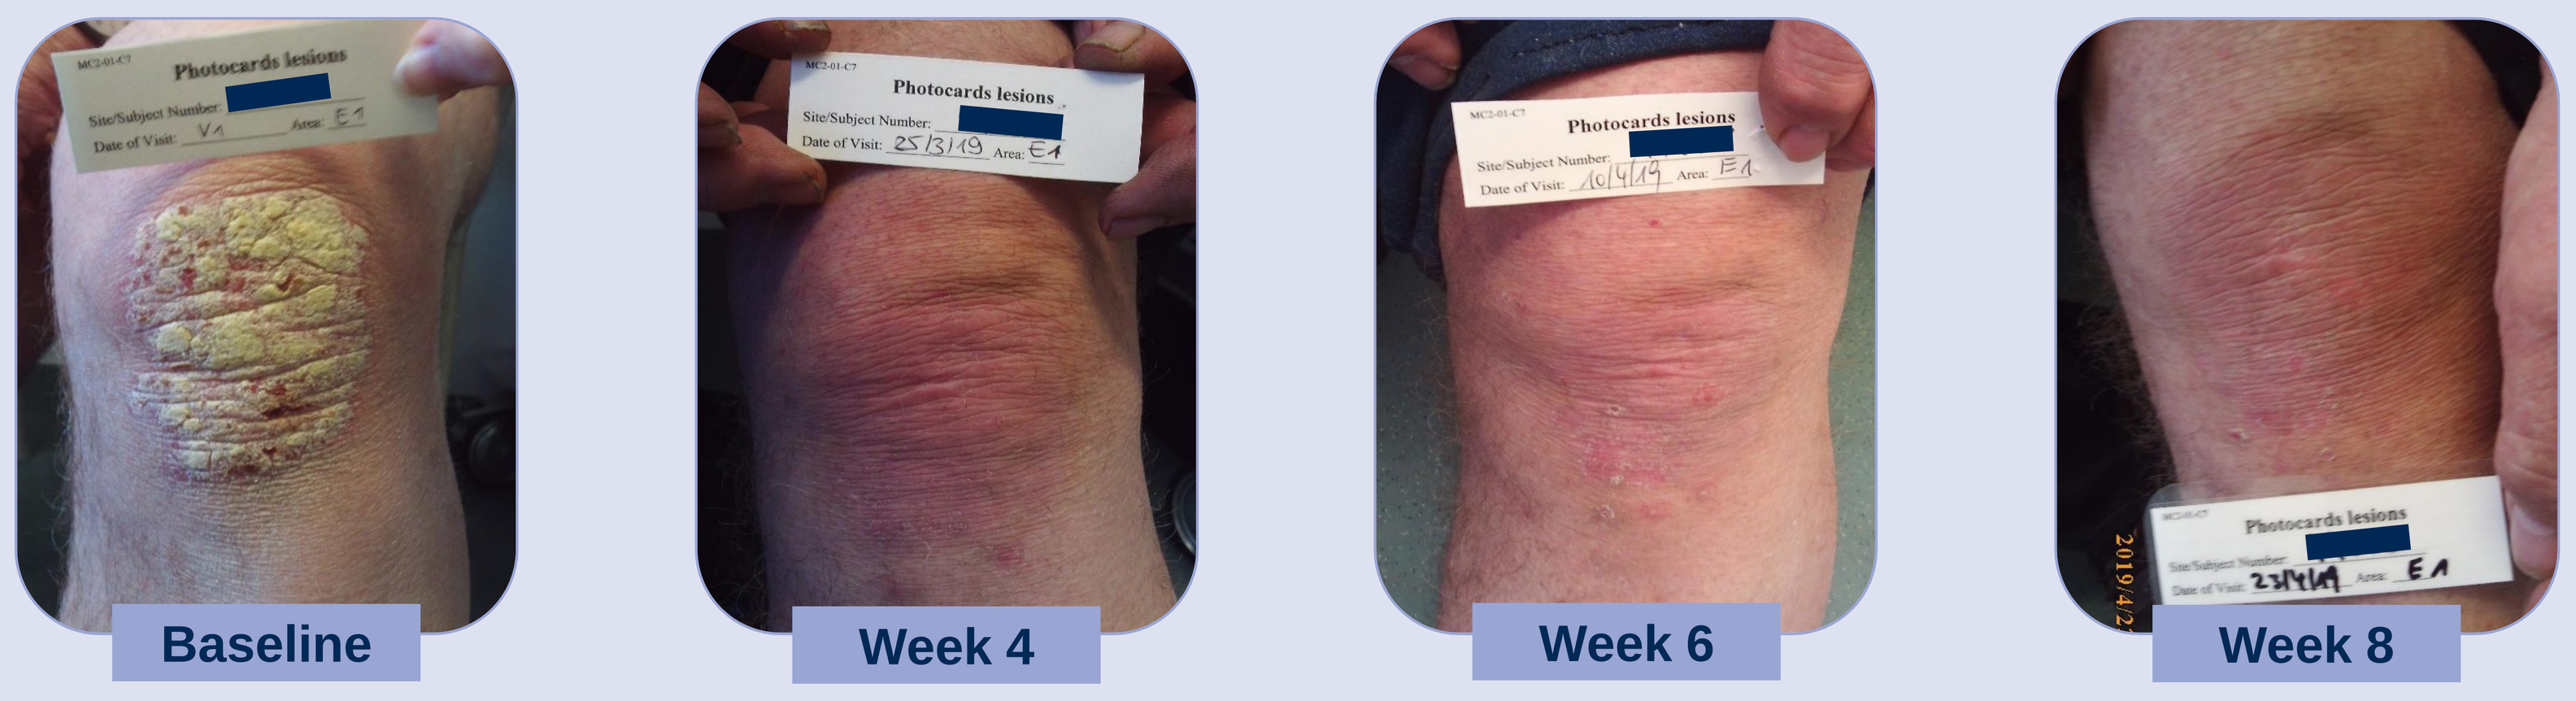
Baseline
Week 4
Week 6
Week 8

CASE 2

Legs: Considerable improvement observed as early as Week 1, with near-complete clearance achieved by Week 8

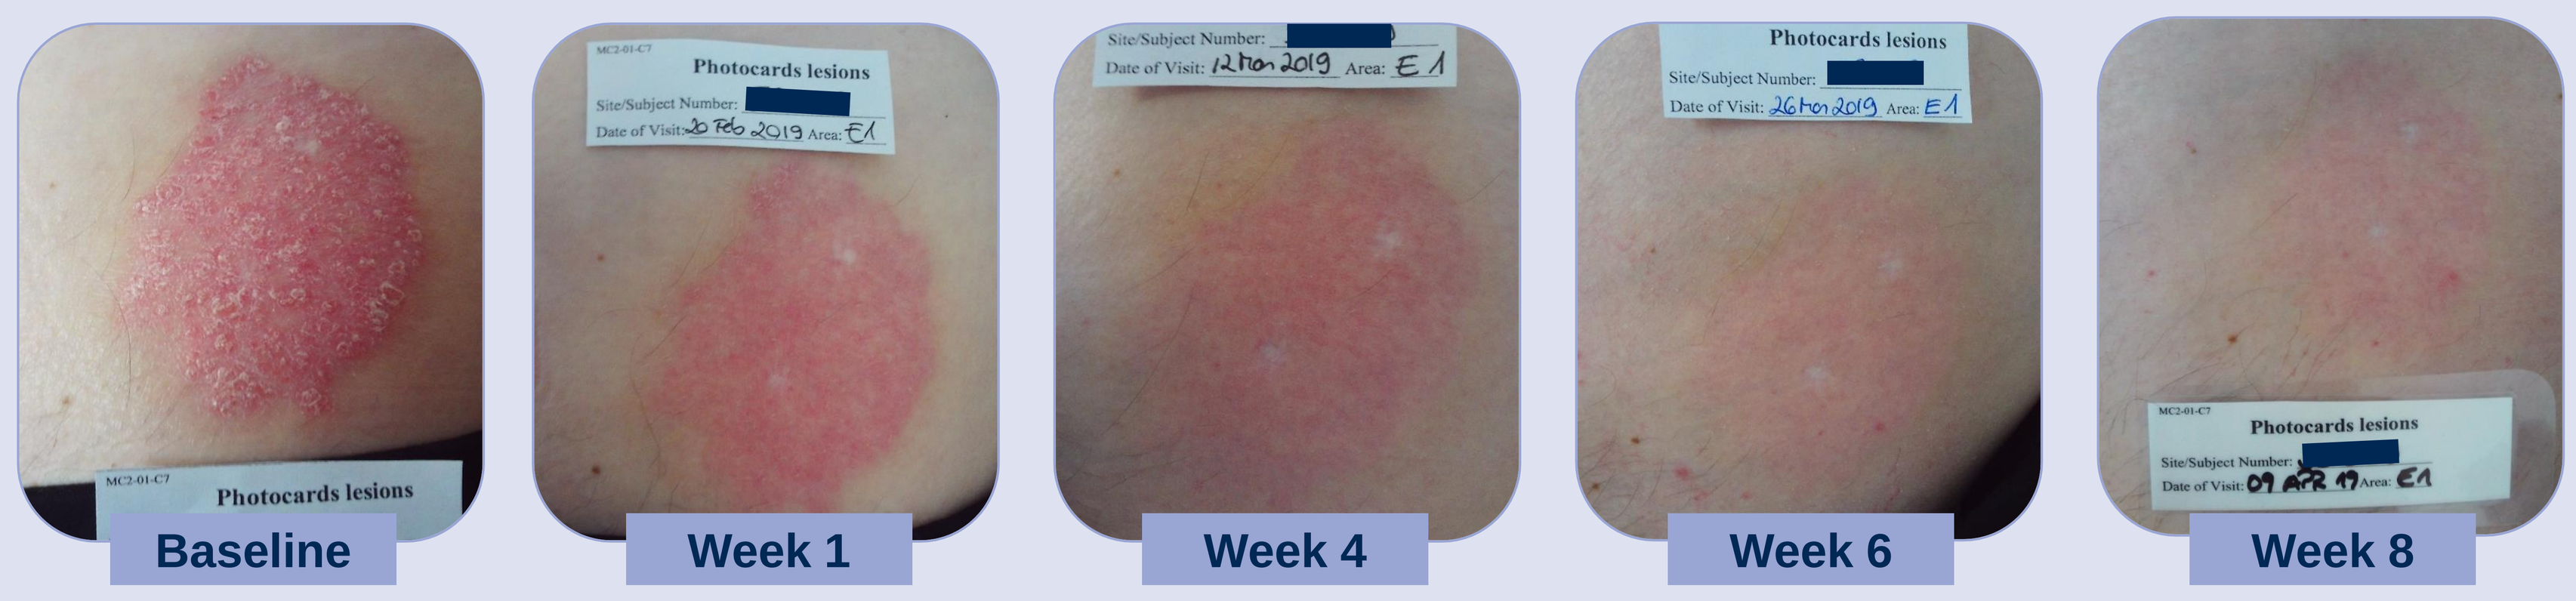
Baseline
Week 1
Week 4
Week 6
Week 8

CASE 3

Buttocks: Favourable progression of erythemato-squamous plaques on the buttocks

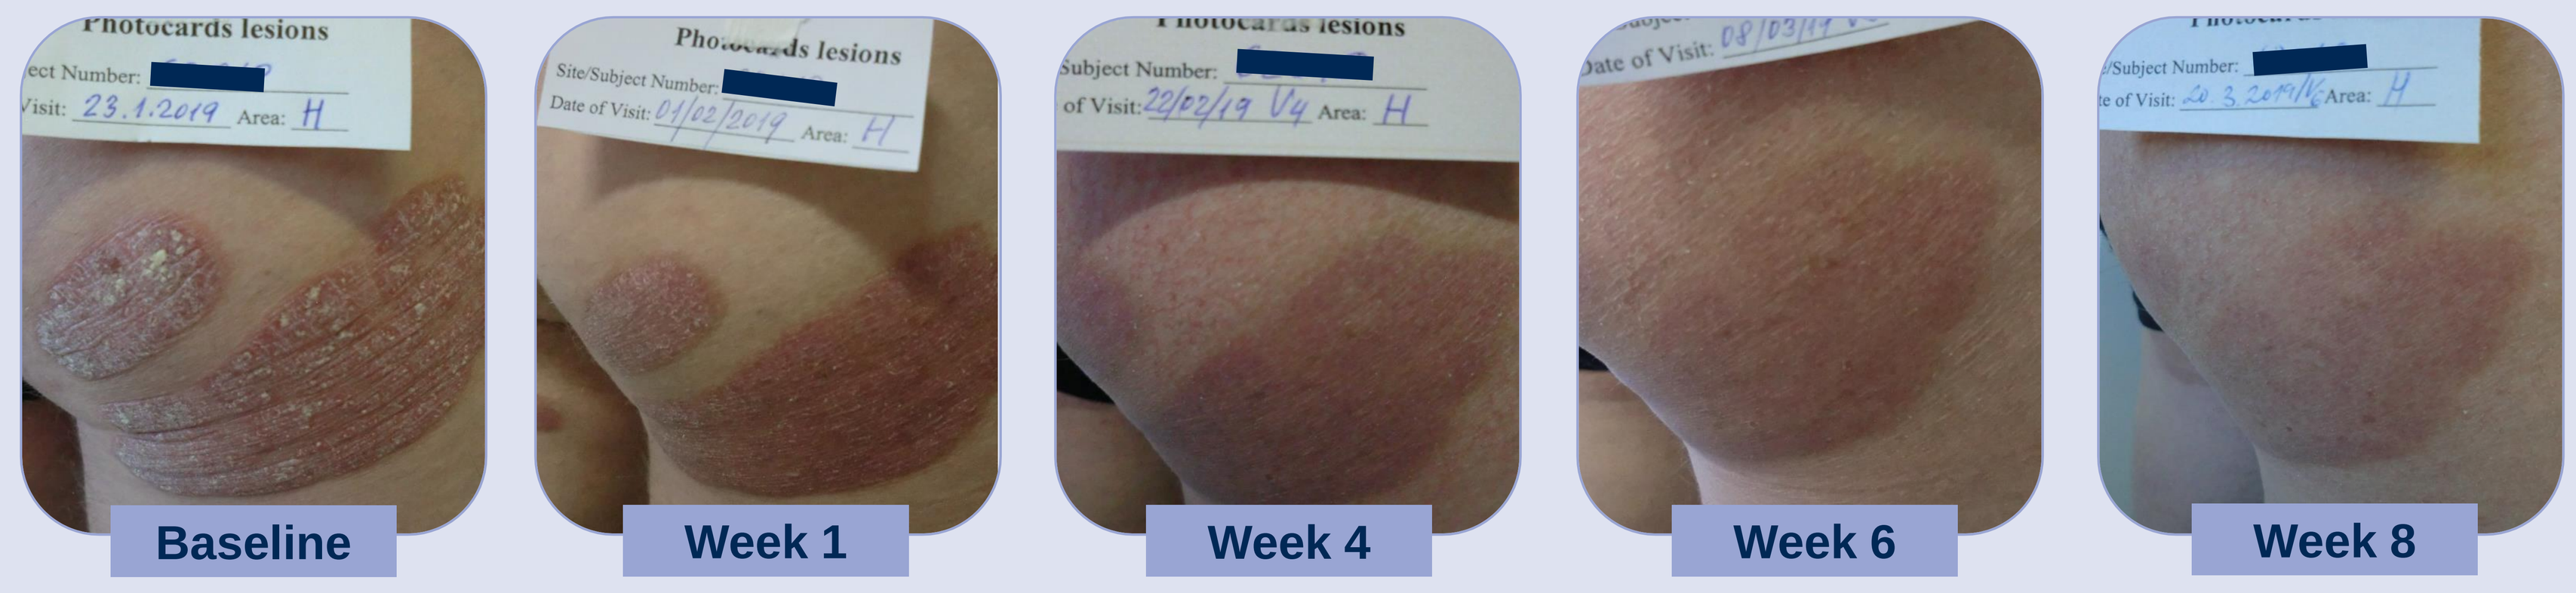
Baseline
Week 1
Week 4
Week 6
Week 8

CASE 4

Elbows: Erythemato-squamous plaques on the elbows. The patient achieved best response already at Week 4

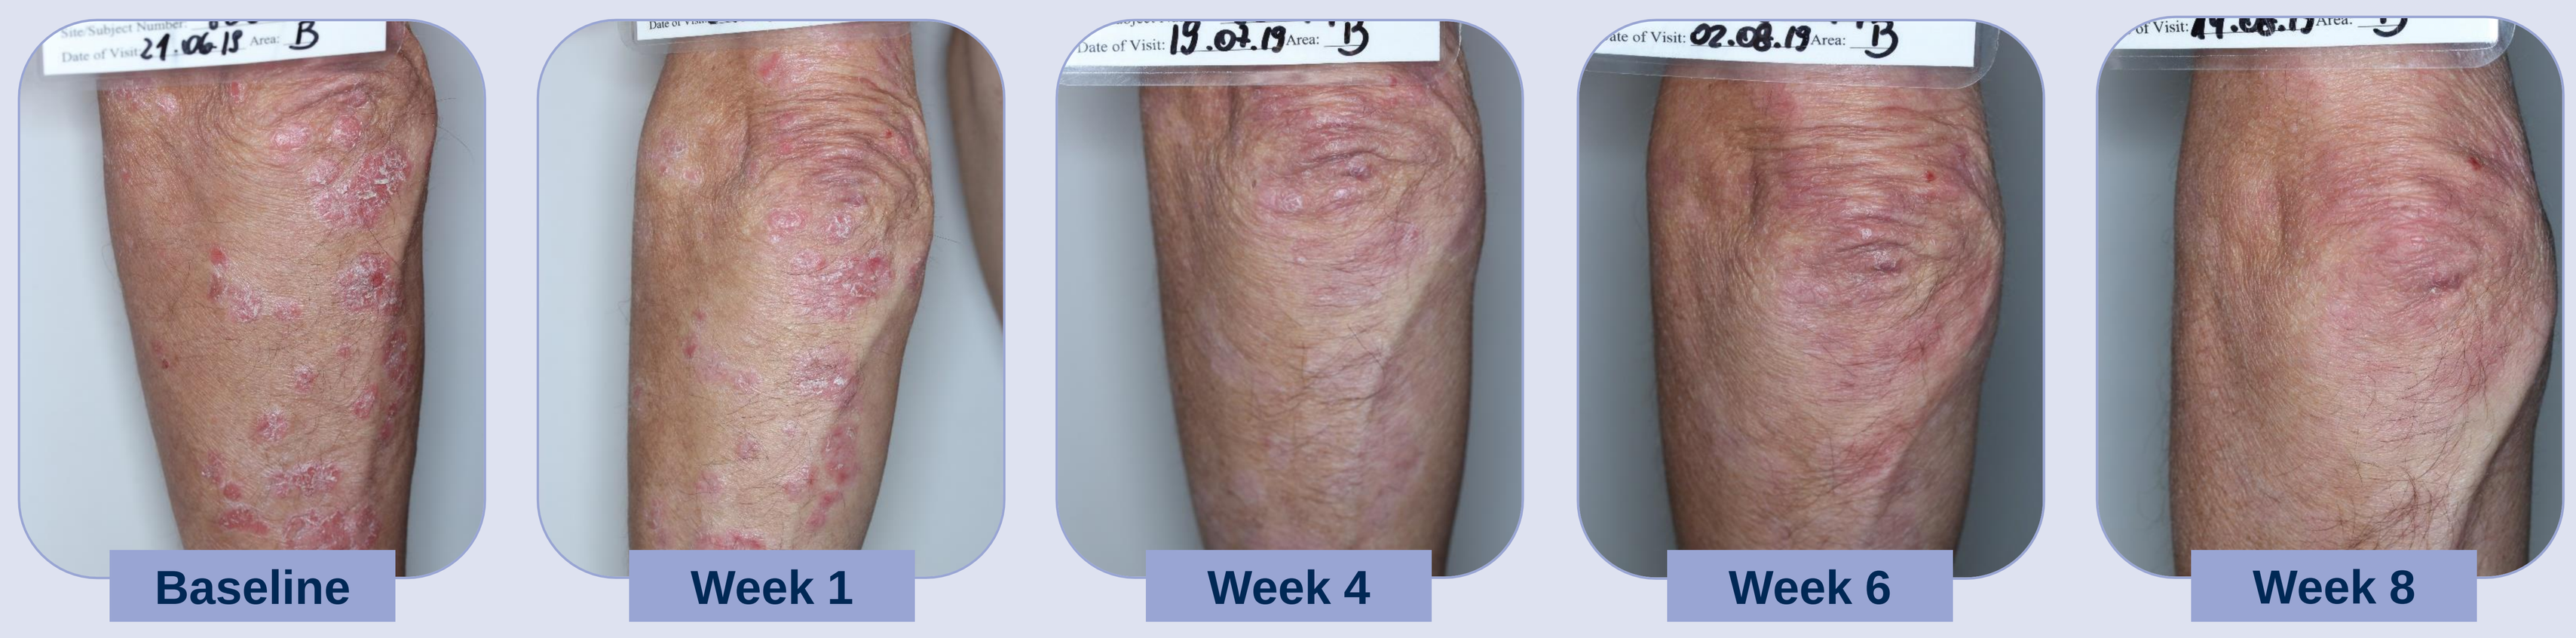
Baseline
Week 1
Week 4
Week 6
Week 8

CASE 5

Trunk: Most of the plaques were nearly healed after 4 weeks of treatment, with only mild residual erythema remaining

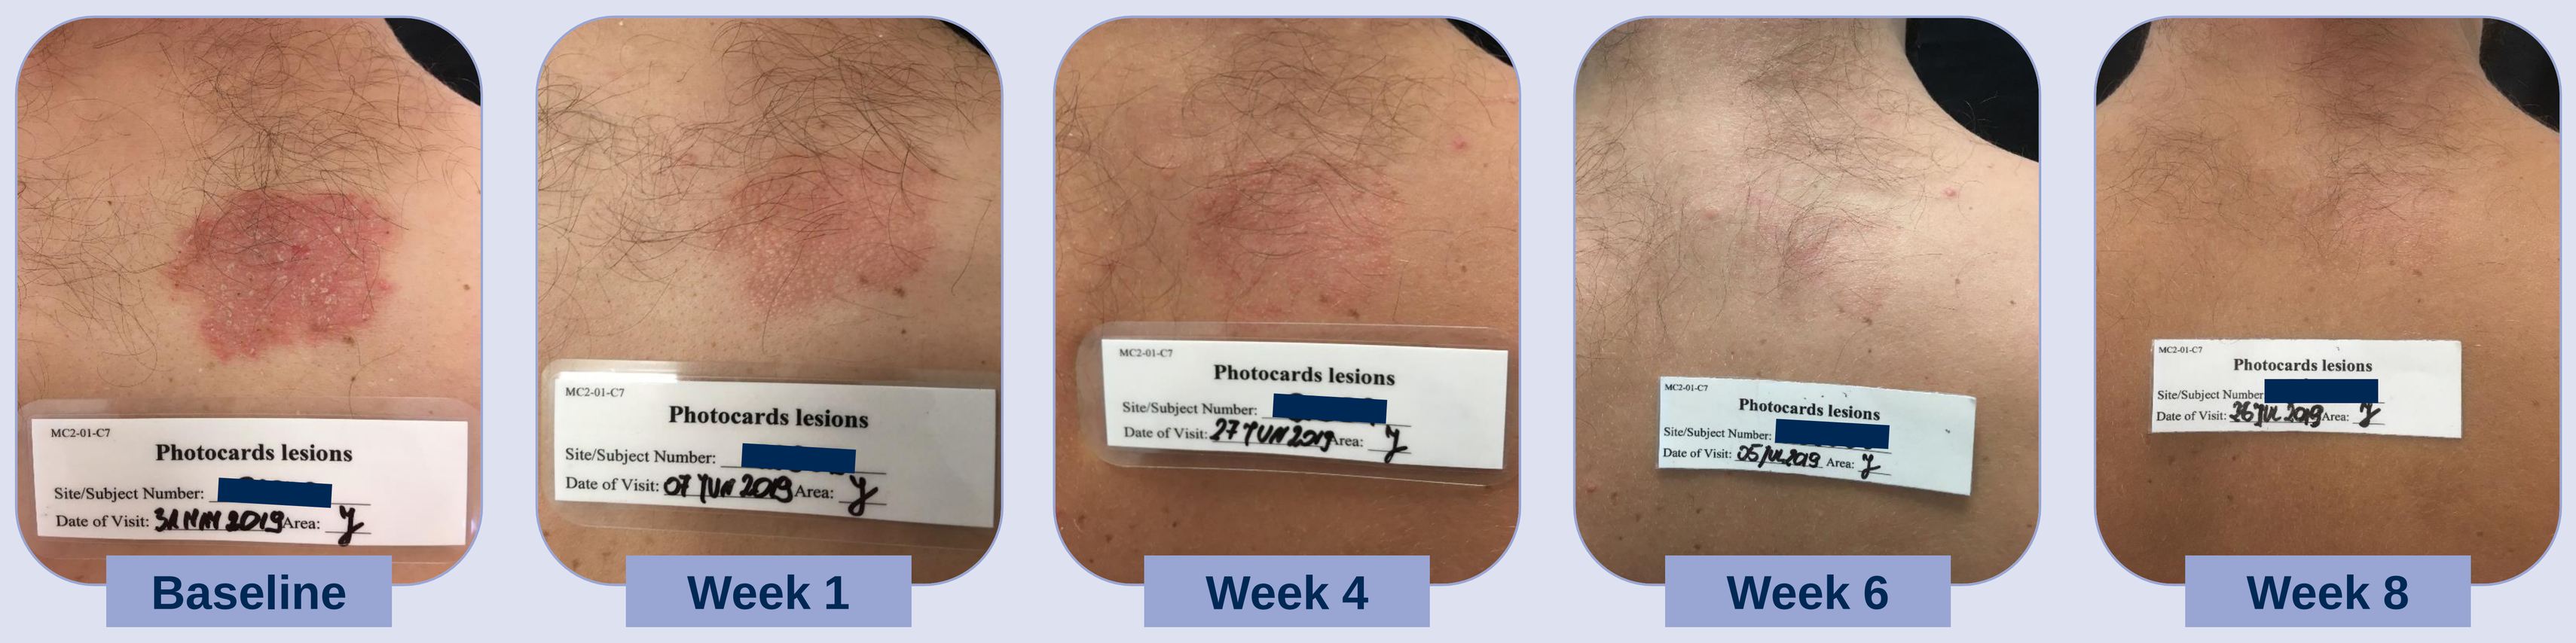
Baseline
Week 1
Week 4
Week 6
Week 8

CONCLUSIONS

- We present five case reports demonstrating the efficacy of CAL/BDP PAD-cream in treating mild-to-moderate psoriasis while improving patients' quality of life.
- Despite the existing variability in key baseline characteristics, this entire group of patients reached the restrictive best response composite endpoint, which combines two efficacy outcomes (PGA and mPASI) and one quality of life outcome (DLQI) after 8 weeks of treatment.
- Furthermore, one case showed an early response after only 4 weeks of treatment with CAL/BDP PAD-cream.

REFERENCES

1. Torres T, et al. *Dermatol Ther (Heidelb)*. 2023;13(10):2153-2169.
2. Pinter A, et al. *J Eur Acad Dermatol Venereol*. 2022;36(2):228-236.
3. Pinter A, et al. *Dermatol Ther (Heidelb)*. 2025;15(6):1441-1453.
4. Pinter A, et al. *J Eur Acad Dermatol Venereol.* 2023; 37(11): 2327–2335.

ACKNOWLEDGEMENTS

- TFS HealthScience provided support in the preparation of this poster.
- This analysis was funded by Almirall.

34th EADV Congress. Paris, 17th-20th September 2025